Predicting Who Will Develop Treatment Resistant Depression after Being Newly Diagnosed With Depression

Jenna Reps, PhD[1], M. Soledad Cepeda MD PhD[1], Patrick B Ryan, PhD[1]

[1]Janssen Research & Development, LLC, Titusville, NJ, USA

BACKGROUND

- No treatment for major depressive disorder is uniformly effective, and subsequent interventions are often needed.
- Patients who do not respond to antidepressants are said to have treatment resistant depression (TRD).
- If one could predict whether a patient who is being diagnosed with depression for the first time would develop TRD, health care providers could monitor these patients more closely or implement treatments in different ways.

PREDICTION PROBLEM

Amongst a target population of newly diagnosed depressed people with therapeutic treatment, who will develop the outcome of TRD within a year of the start of depression diagnosis?

METHOD

The analysis used the OHDSI standardised PatientLevelPrediction framework and software [1].

- **Target Population:** People age 18 or older with a first time prescription of an antidepressant, 365+ days of prior observation, no history of psychosis and first record of depression occurring within 60 days prior to the antidepressant. The target population cohort start date was the date of the first antidepressant.
- **Outcome:** Three or more distinct antidepressants or an antidepressant and antipsychotic during the time at risk.
- **Time-at-risk:** From 0 days until 365 days after the target population cohort start date (the date of the first antidepressant).
- **Variables:** All conditions, drugs, procedures, measurements and observations in the 365 days prior to target cohort start date, in addition to demographic such as age and gender.
- **Model:** A regularised lasso logistic regression model was trained due to the large number of variables,
- **Training:** 75% of the data from a USA claims dataset (Truven Commercial Claims and Encounters (CCAE), were used to trained the model, with 3-fold cross validation implemented to identify the optimal hyper-parameters,
- **Internal Validation:** The remaining 25% of the data were used to validate the performance. Area under the receiver operating characteristic curve (AUC) where used to determine discriminative ability and calibration plots were inspected to determine model calibration.

RESULTS

There were 370,322 people in the target population but 139,521 of the target population were censored, leaving 230,801 people. Within the target population, 24037 people had TDR within a year (6.5% of the original population and 10.4% of the uncensored population had the outcome). Figure 1 shows the characterisation of the outcome vs non-outcome people in the target population.

Figure 1: **Variable incidence comparison between people with and without the outcome.**

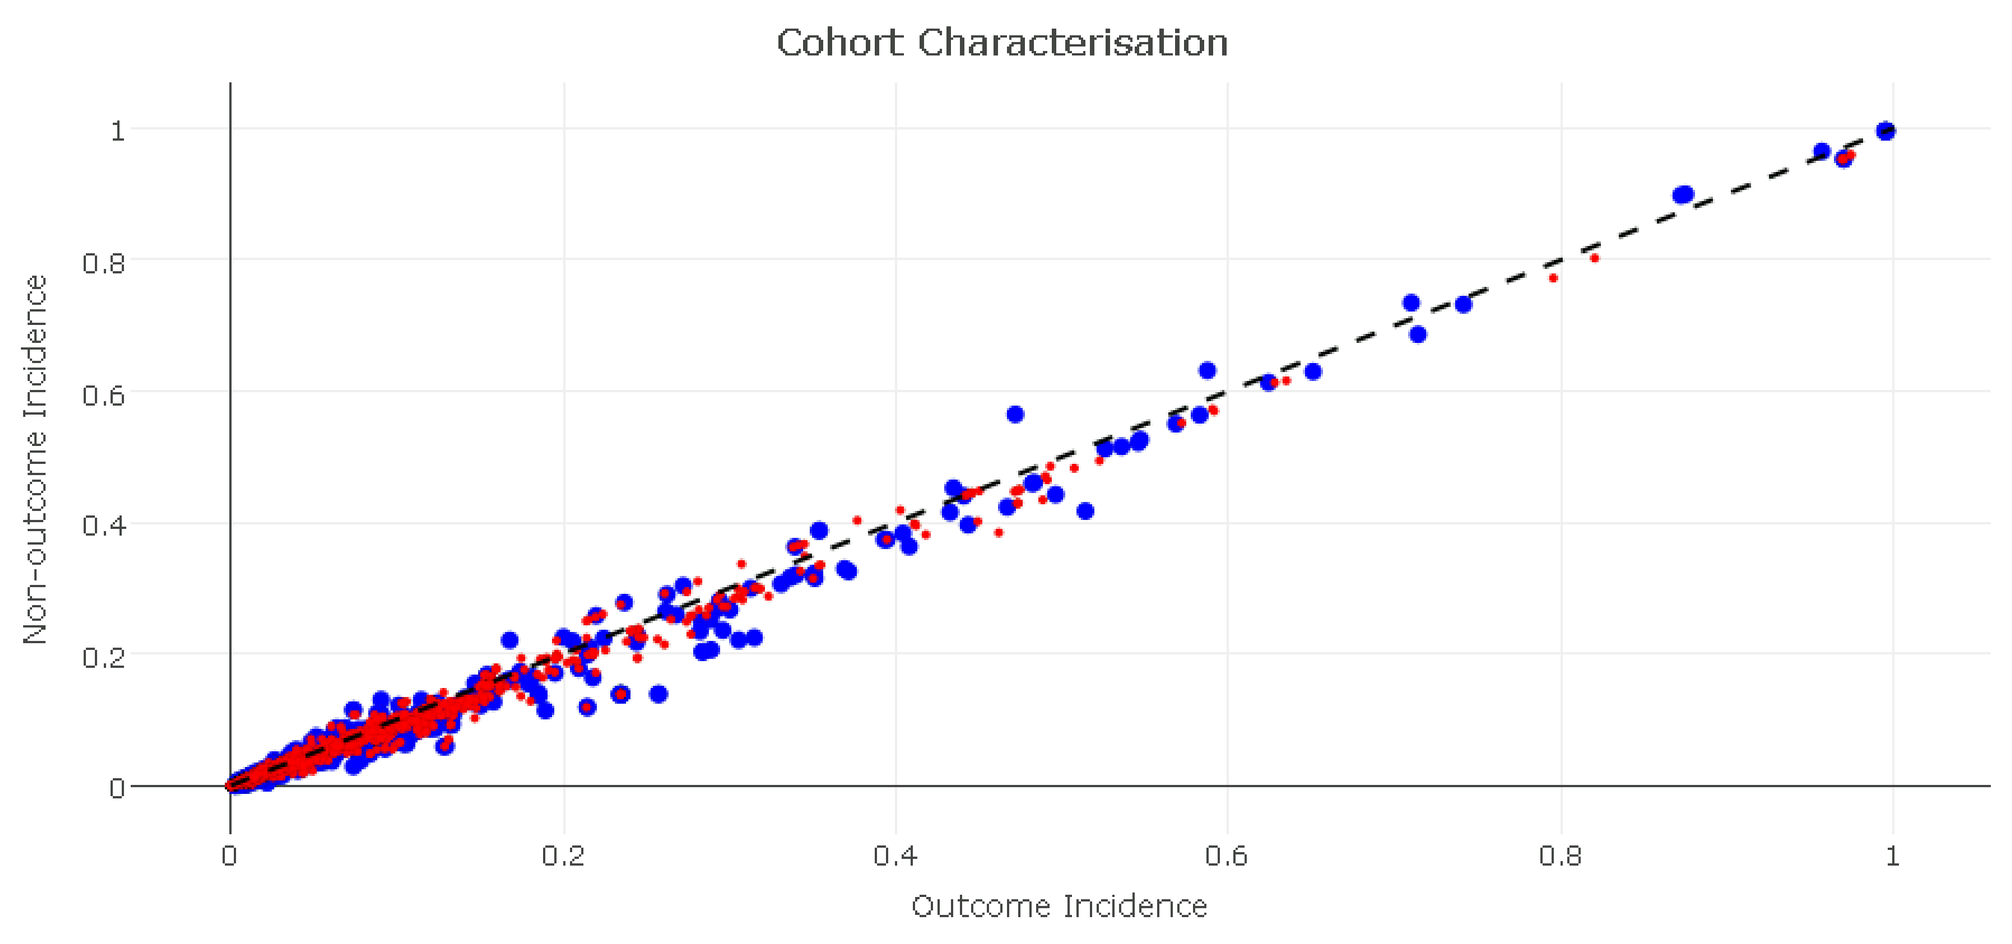
Cohort Characterisation
Non-outcome Incidence
Outcome Incidence

Figure 2: **ROC plot for the test set**

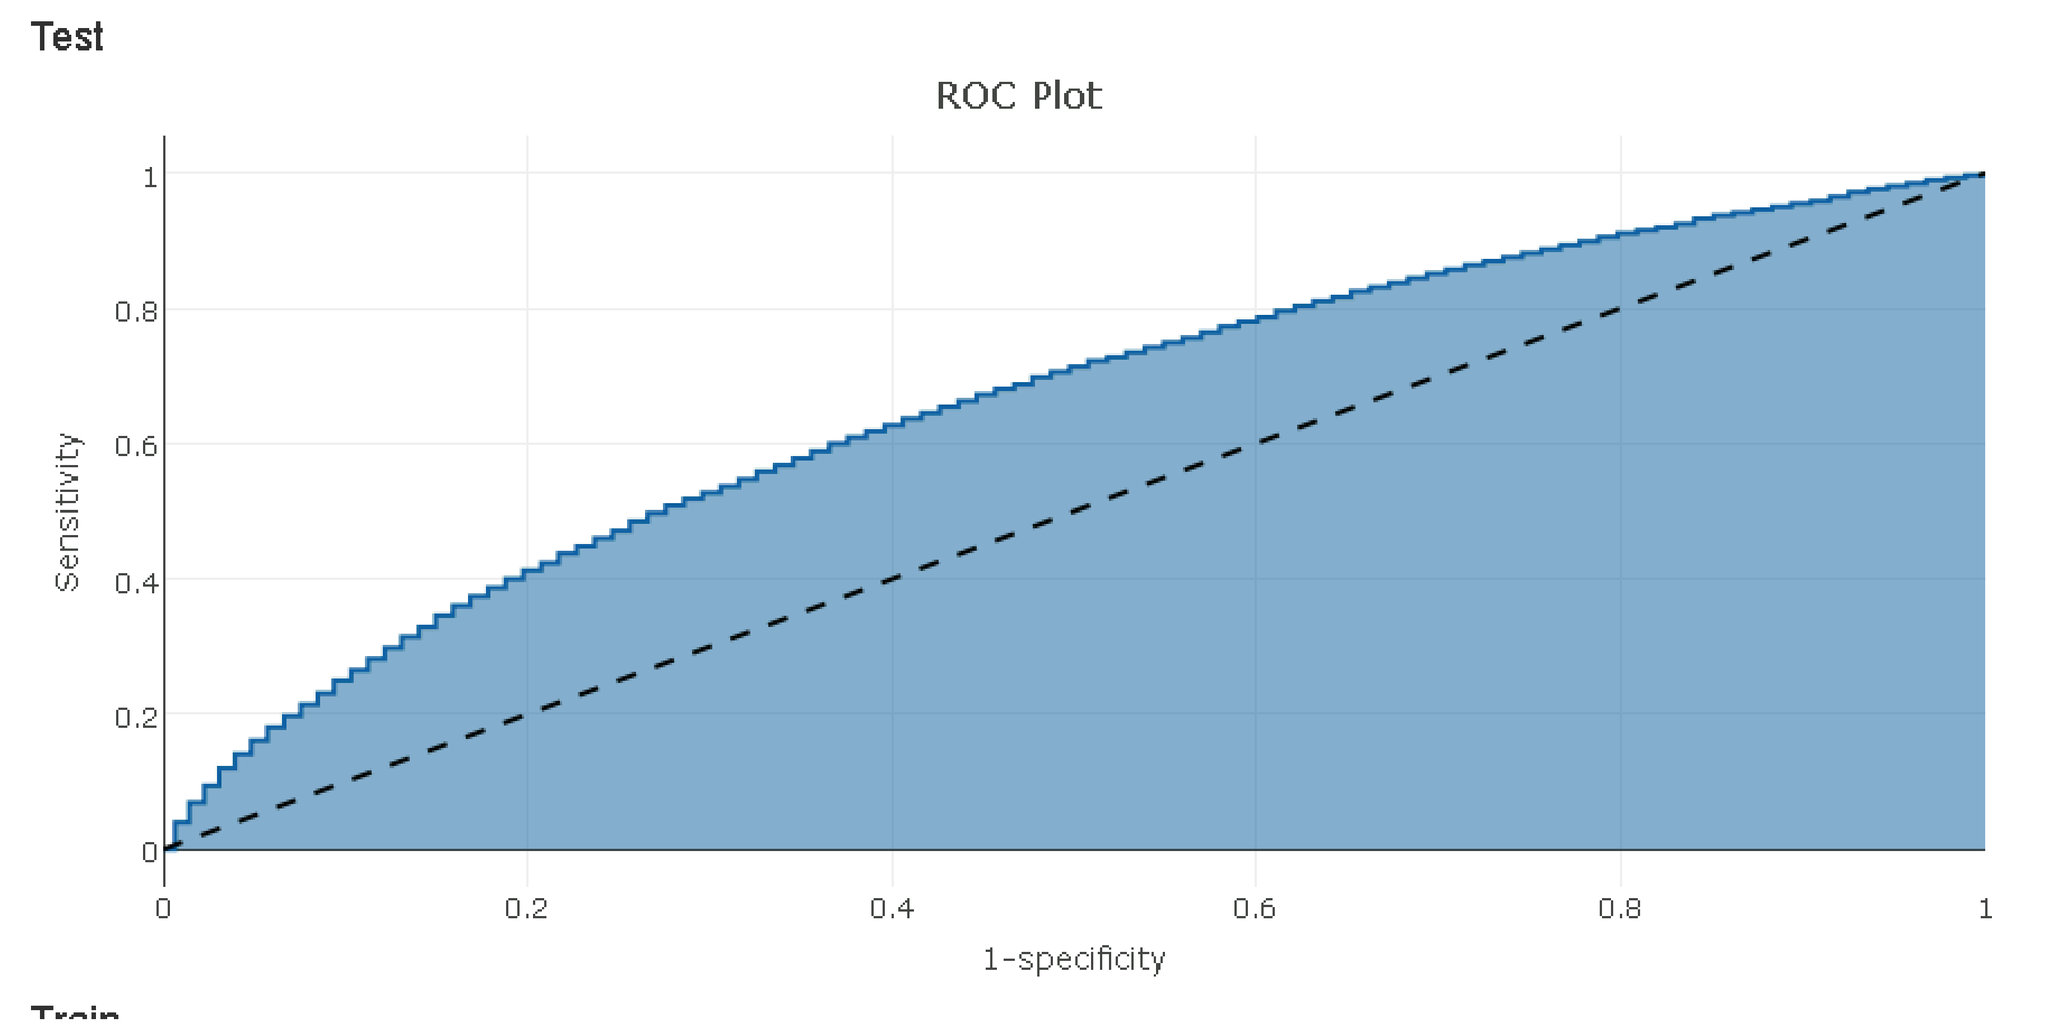
Test
ROC Plot
Sensitivity
1-specificity

The model obtained an internal validation of 0.66 (0.65.-0.67.), see figure 2 for the ROC plot. The model was well calibrated as the predicted risk matched the observed risk for the different risk bins, see figure 4. It was also well calibrated across age and gender groups, see Figure 4.

Figure 3: **Calibration plot for the test set**

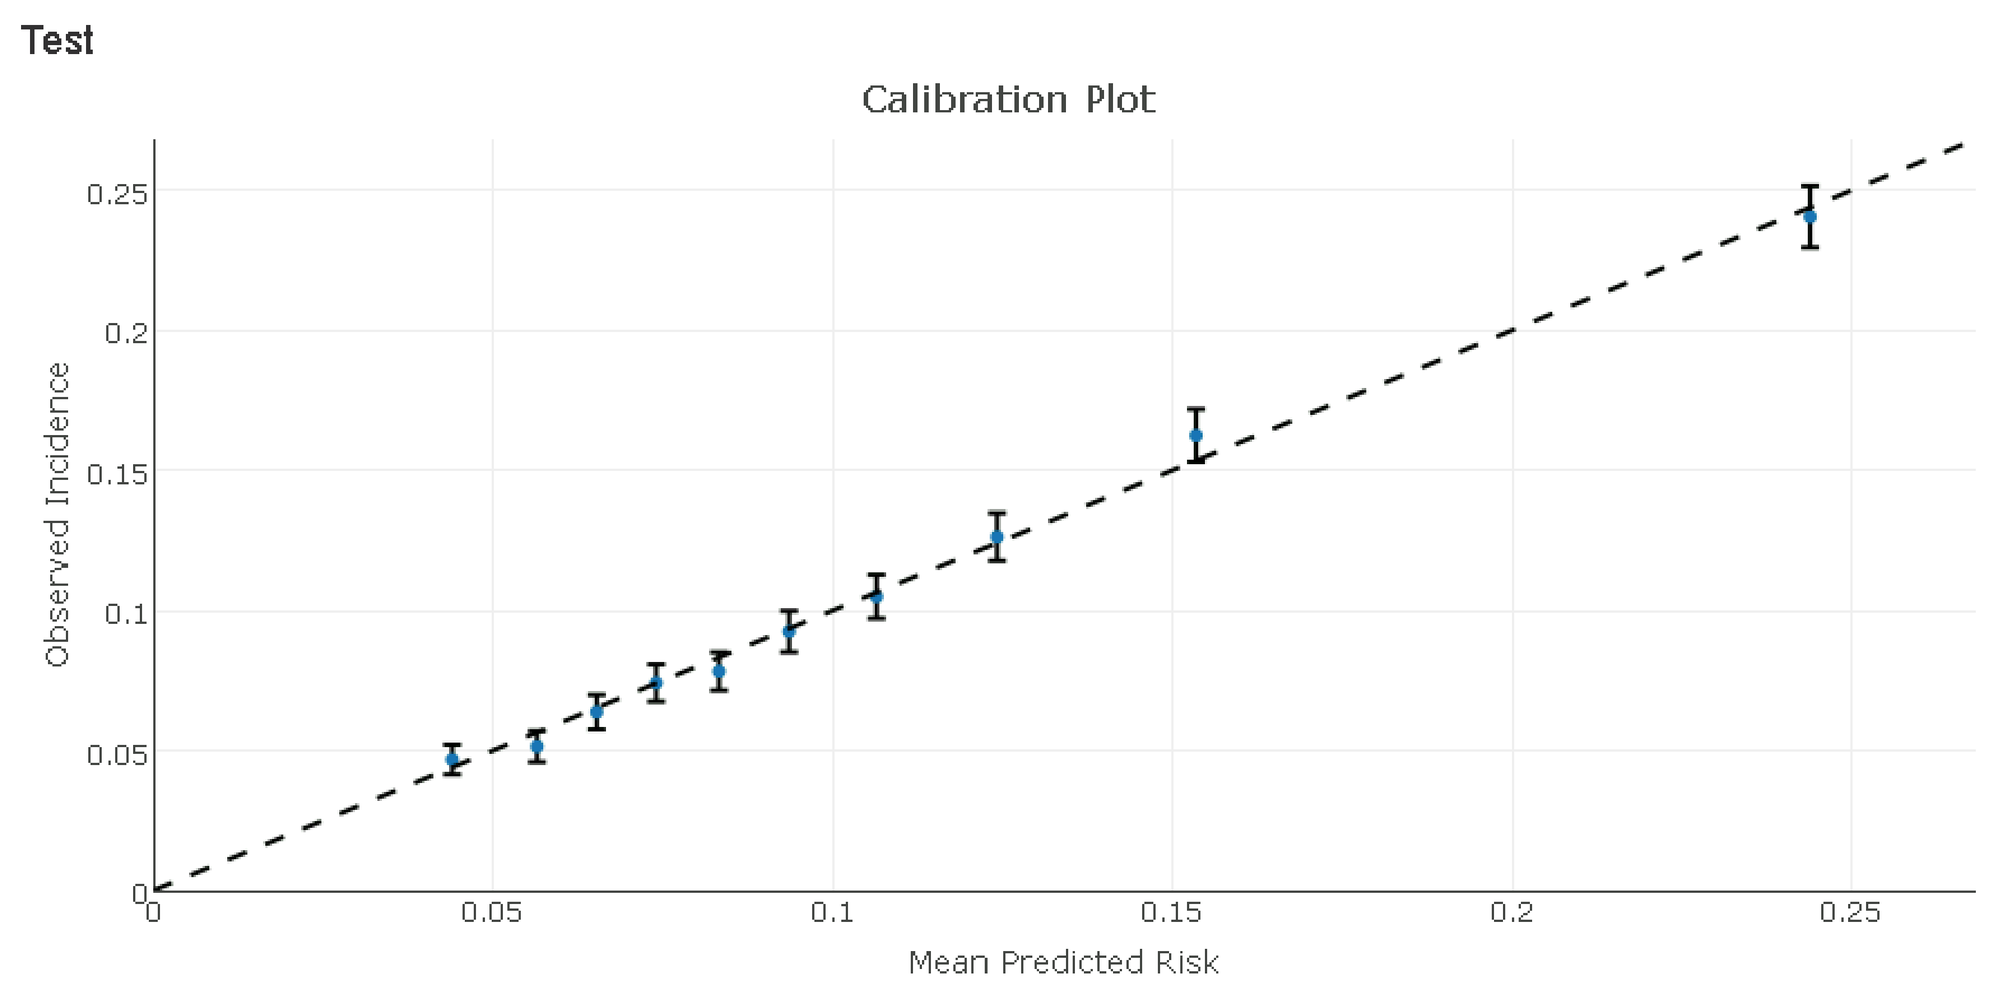
Test
Calibration Plot
Observed Incidence
Mean Predicted Risk

Figure 4: **Demographic calibration**

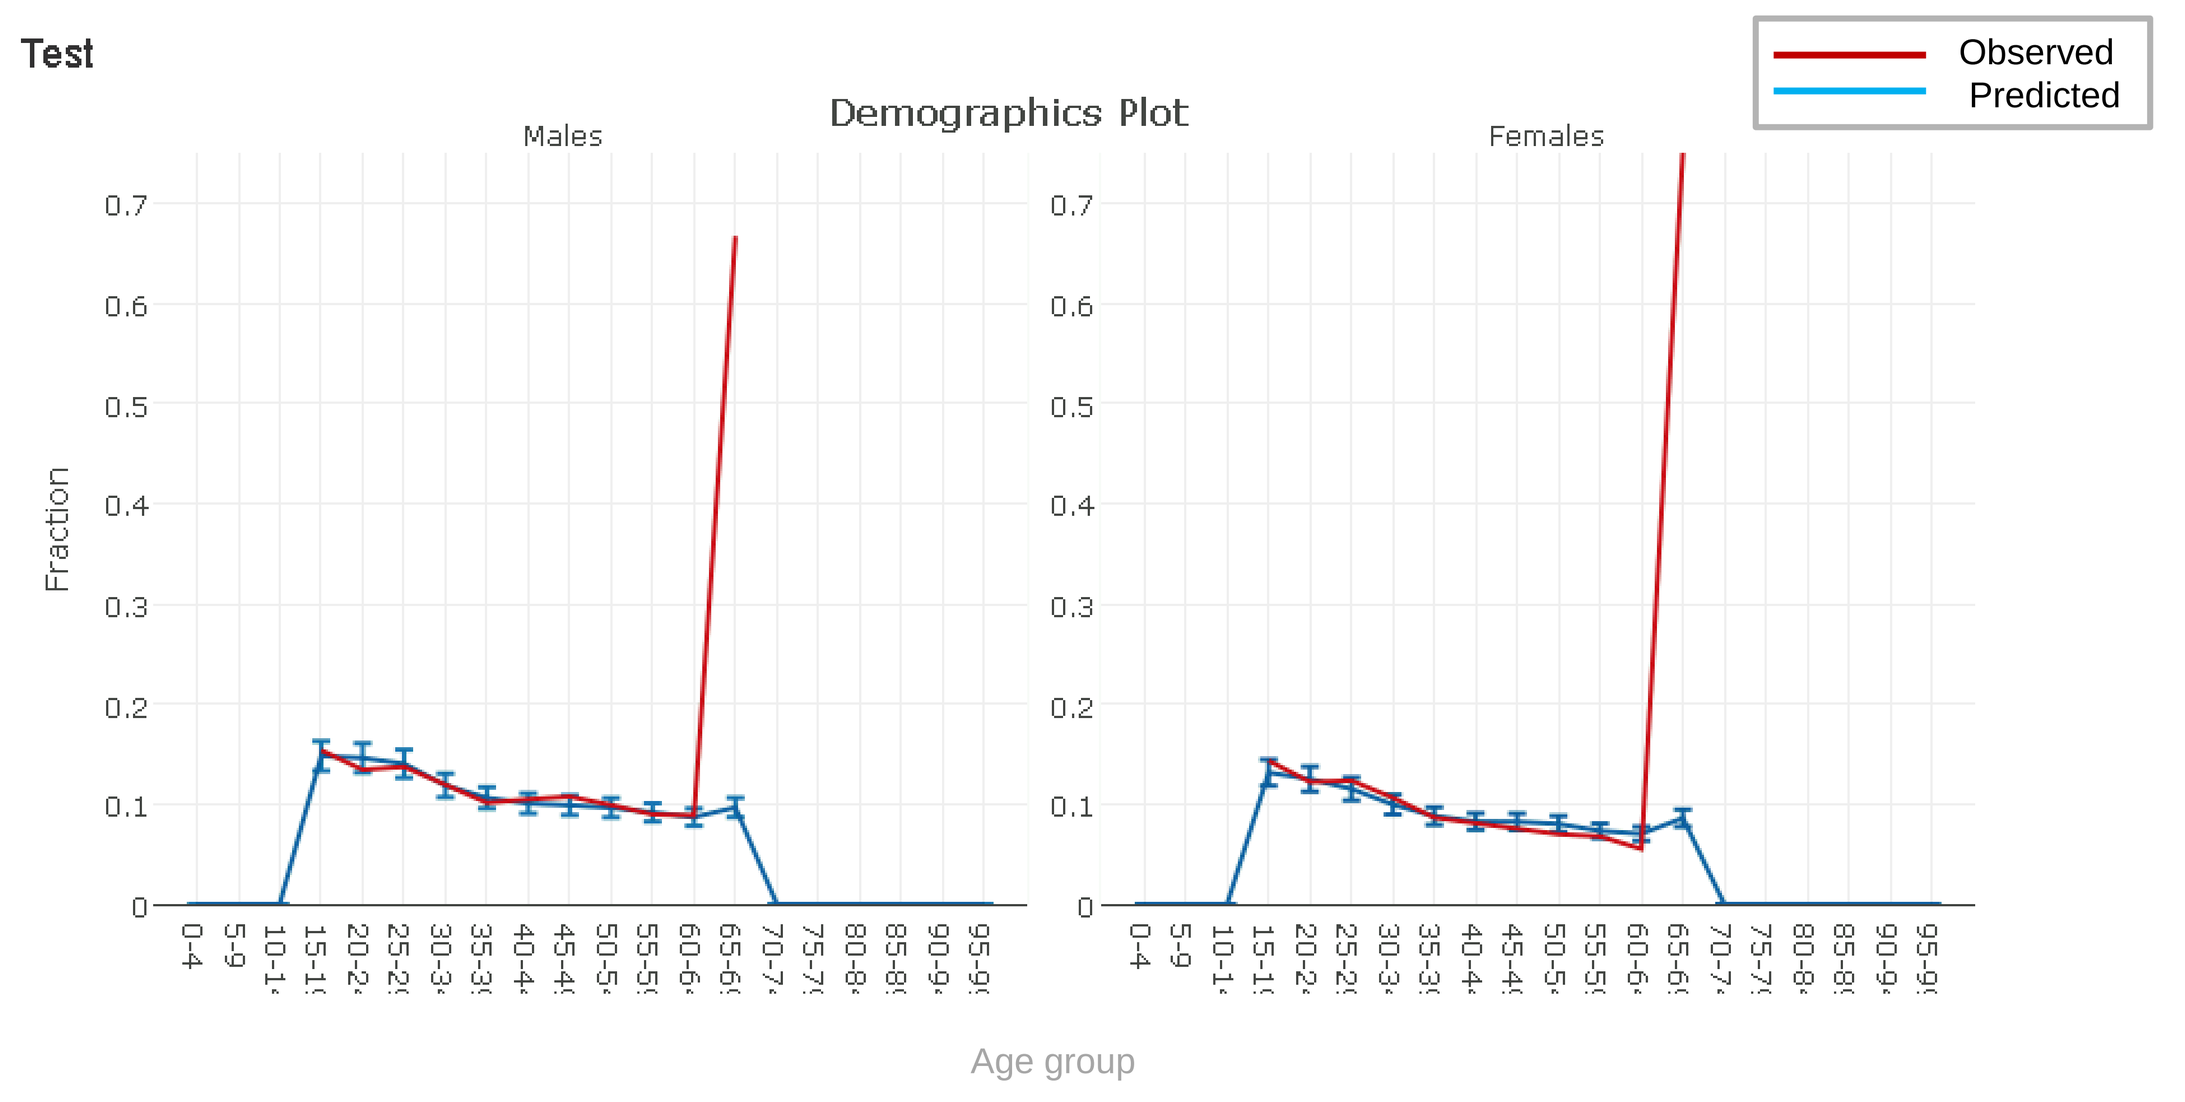
Test
Demographics Plot
Males
Females
Observed
Predicted
Fraction
Age group

It was found that TRD patients at baseline were younger, 10.87% were between 18 to 19 years old versus 7.64% in the no TRD group, RR=1.42, (95% CI 1.37-1.48). TRD patients were more likely to have a diagnosed anxiety disorder at baseline than non-TRD patients, RR=1.38, (95% CI 1.35-1.14). Fatigue, had the highest risk ratio, RR= 3.68, (95% CI 3.18-4.25). TRD patients also had diagnosed substance use disorders, psychiatric conditions, insomnia and pain more often at baseline than non-TRD patients.

CONCLUSION

- Ten percent of the subjects newly diagnosed and treated for depression developed TRD within a year. They were younger and suffered more frequently from fatigue, substance use disorders, anxiety, psychiatric conditions, insomnia and pain than non-TRD patients.
- The discriminate performance of the model was modest and in future work it would be useful the test more advanced machine learning techniques such as deep learning with the aim of developing models with higher discriminative ability.

Conflict of Interest Statement

All authors are employees of Janssen Research & Development, LLC. Janssen Research & Development, LLC has an interest in depression and treatment resistant depression

References: Jenna Reps, Martijn J. Schuemie, Marc A. Suchard, Patrick B. Ryan and Peter R. Rijnbeek (2017). PatientLevelPrediction: Package for patient level prediction using data in the OMOP Common Data Model. R package version 1.2.1.