Cardio- and cerebrovascular risk in users of conventional synthetic disease-modifying antirheumatic drugs (csDMARDs) for rheumatoid arthritis (RA)

SAT0131

Albert Prats-Uribe (1); Ben Illigens (2, 3); David Vizcaya (4); James Weaver (5); Edward Burn (1, 6); Patrick Ryan (5); Ruta Sawant (7); Daniel Prieto-Alhambra (1, 6); and the EHDEN RA Research Group

1 - NDORMS, University of Oxford;
2 - Beth Israel Deaconess Medical Center, Department of Neurology, Harvard Medical School;
3 - Odysseus Inc.; 4 - Bayer Hispania, Spain;
5 - Janssen Research and Development; 6- IDIAPJGol, Spain ;
7 - Abbvie

More information, protocol, code and detailed results from this study scanning the QR code or at: https://data.ohdsi.org/EhdenRaDmardsEstimation/

Background

Rheumatoid arthritis (RA) is associated with an up to 60% increased risk of cardio- and cerebrovascular disease (CVD). csDMARDs used as first-line treatments for RA can potentially mitigate this risk but limited data are available on their comparative safety effects. Previous trials have reported protective effects for methotrexate (MTX) and recently hydroxychloroquine (HCQ), but no trial to our knowledge has shown such benefits for sulfasalazine (SSZ) or leflunomide (LEF).We aimed to assess the comparative safety effect of most common first-line csDMARDS on the risk of myocardial infarction (MI) and stroke (ischemic and hemorrhagic) in RA patients.

Methods

The study has been informed by data from 6 databases from Germany, US, and UK, all mapped to a common data model. The multinational real world cohort study was conducted including patients who were ≥18 years old, had first recorded RA diagnosis between 2005-2019, were csDMARD naive and initiated a monotherapy with MTX, HCQ, SSZ, or LEF. Those with a prior diagnosis of other inflammatory arthropathy were excluded. Patients were followed until the date of first outcome event, death, loss of or end to follow-up (5 years).

Figure 1. Calibrated hazard ratios (cHRs) for MI and stroke

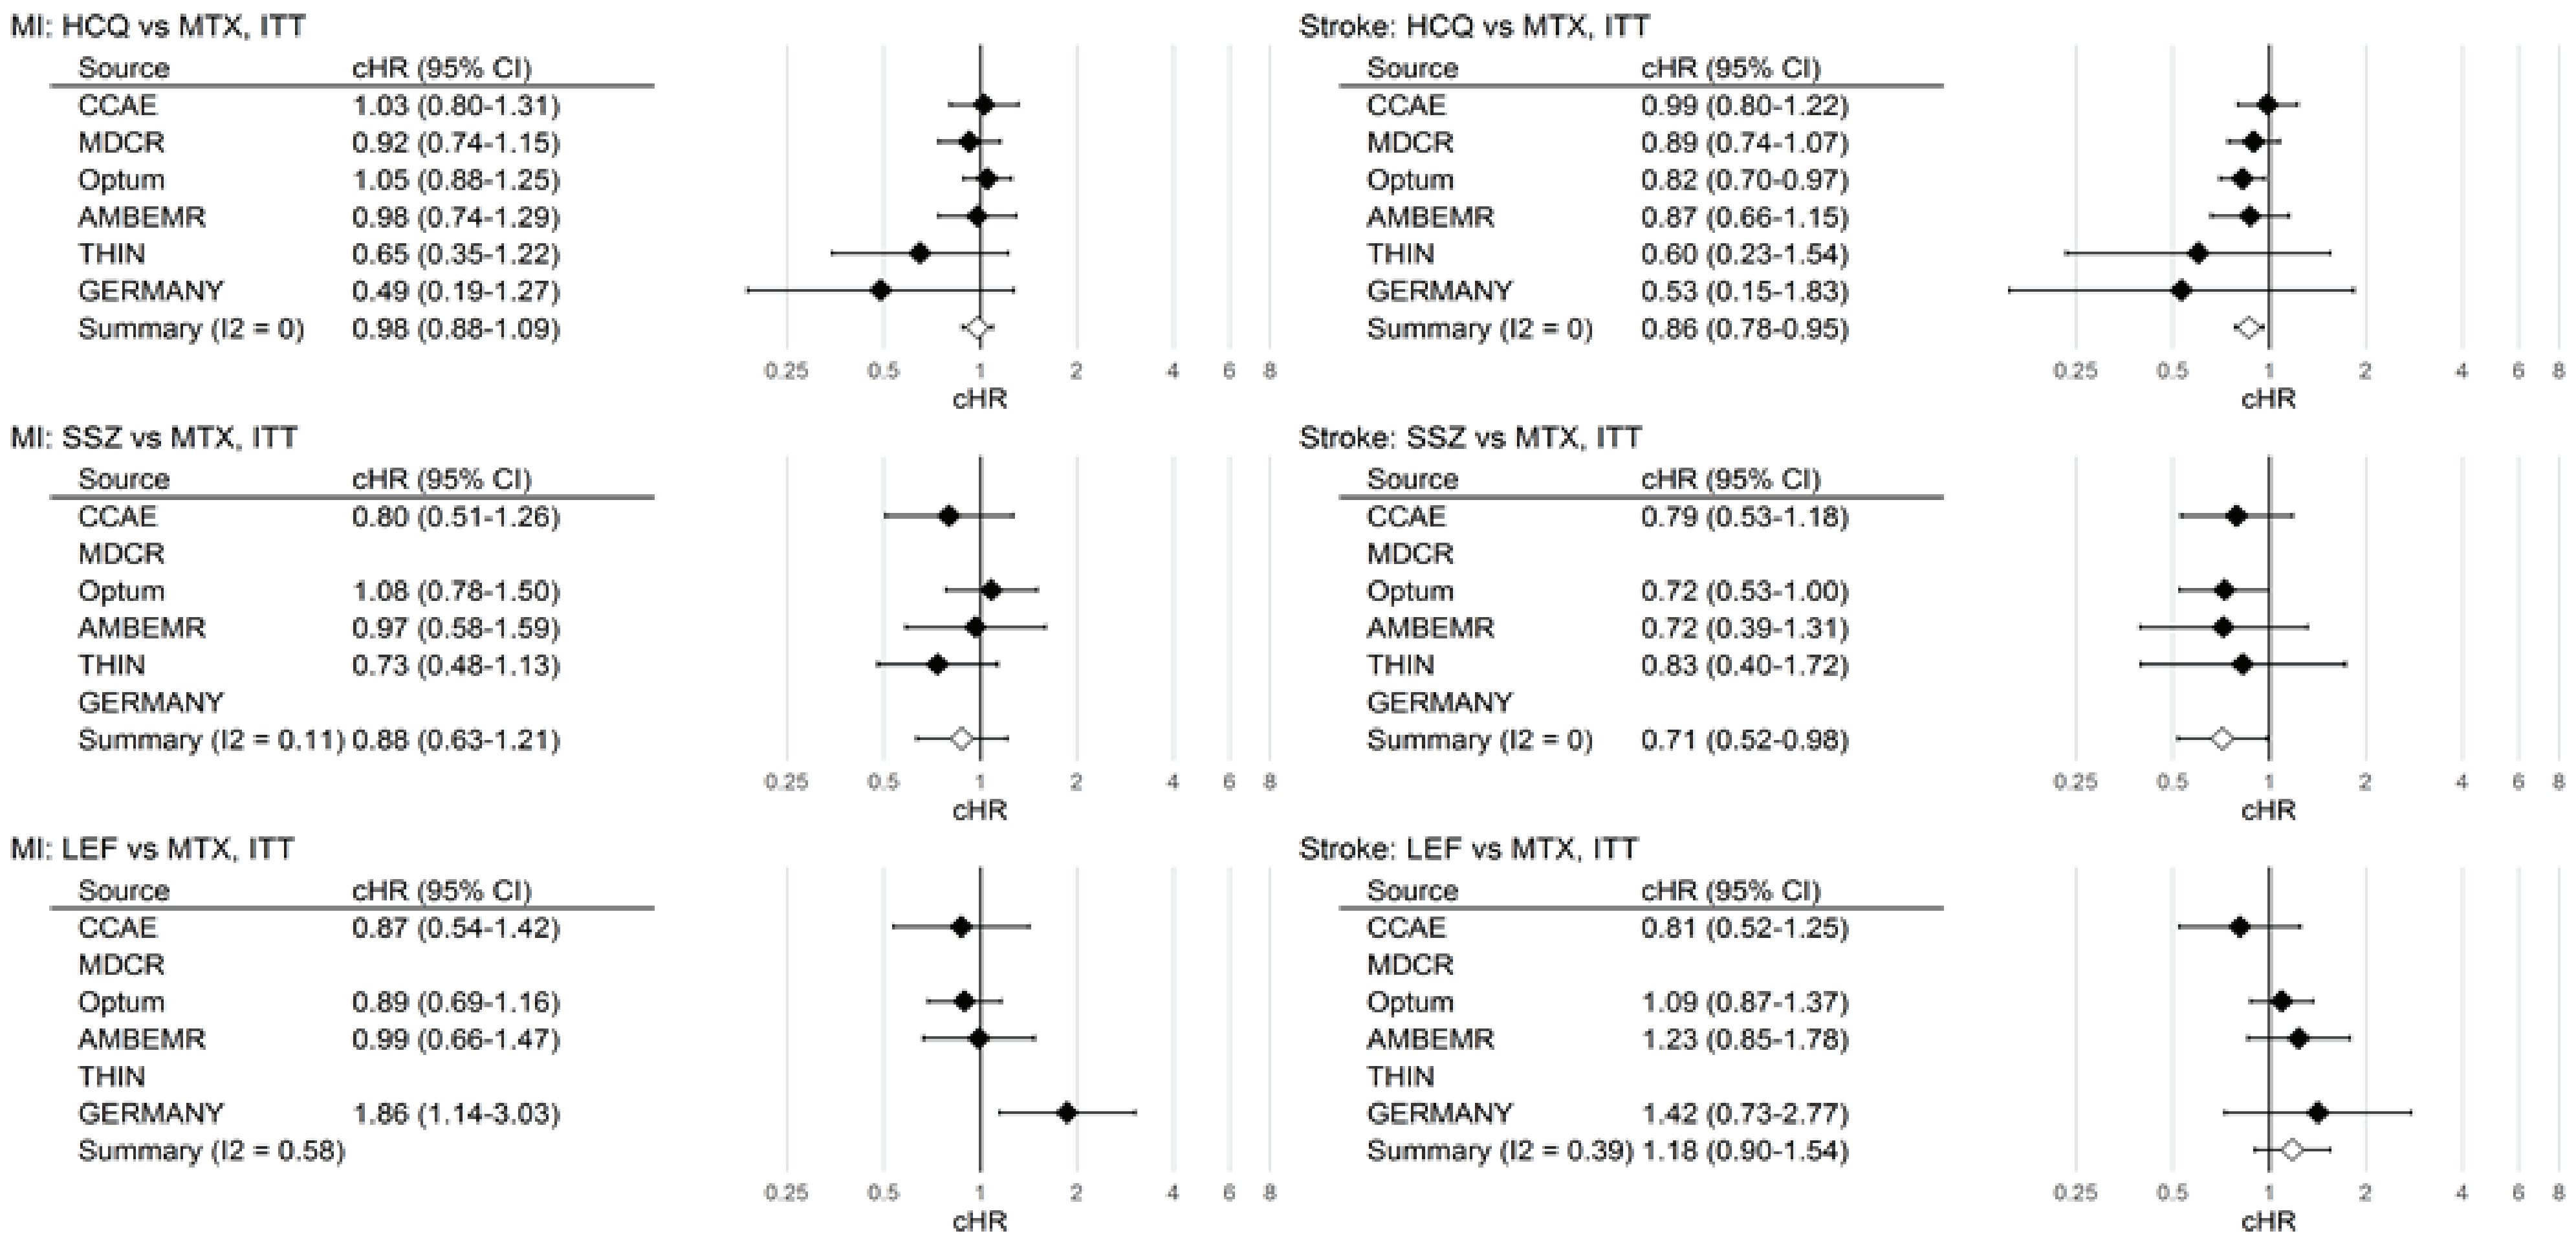

MI: HCQ vs MTX, ITT

Source	cHR (95% CI)
CCAE	1.03 (0.80-1.31)
MDCR	0.92 (0.74-1.15)
Optum	1.05 (0.88-1.25)
AMBEMR	0.98 (0.74-1.29)
THIN	0.65 (0.35-1.22)
GERMANY	0.49 (0.19-1.27)
Summary (I2 = 0)	0.98 (0.88-1.09)

Stroke: HCQ vs MTX, ITT

Source	cHR (95% CI)
CCAE	0.99 (0.80-1.22)
MDCR	0.89 (0.74-1.07)
Optum	0.82 (0.70-0.97)
AMBEMR	0.87 (0.66-1.15)
THIN	0.60 (0.23-1.54)
GERMANY	0.53 (0.15-1.83)
Summary (I2 = 0)	0.86 (0.78-0.95)

MI: SSZ vs MTX, ITT

Source	cHR (95% CI)
CCAE	0.80 (0.51-1.26)
MDCR	
Optum	1.08 (0.78-1.50)
AMBEMR	0.97 (0.58-1.59)
THIN	0.73 (0.48-1.13)
GERMANY	
Summary (I2 = 0.11)	0.88 (0.63-1.21)

Stroke: SSZ vs MTX, ITT

Source	cHR (95% CI)
CCAE	0.79 (0.53-1.18)
MDCR	
Optum	0.72 (0.53-1.00)
AMBEMR	0.72 (0.39-1.31)
THIN	0.83 (0.40-1.72)
GERMANY	
Summary (I2 = 0)	0.71 (0.52-0.98)

MI: LEF vs MTX, ITT

Source	cHR (95% CI)
CCAE	0.87 (0.54-1.42)
MDCR	
Optum	0.89 (0.69-1.16)
AMBEMR	0.99 (0.66-1.47)
THIN	
GERMANY	1.86 (1.14-3.03)
Summary (I2 = 0.58)	

Stroke: LEF vs MTX, ITT

Source	cHR (95% CI)
CCAE	0.81 (0.52-1.25)
MDCR	
Optum	1.09 (0.87-1.37)
AMBEMR	1.23 (0.85-1.78)
THIN	
GERMANY	1.42 (0.73-2.77)
Summary (I2 = 0.39)	1.18 (0.90-1.54)

Database estimates not reported where adequate covariate balance not attained.
Meta-analysis results not reported where I2>0.4.
MEDICARE did not pass diagnostics for SSZ and LEF analyses.
CI: Confidence Interval; MTX: Methotrexate; HCQ: Hydroxychloroquine; SSZ: sulphasalazine; LEF: Leflunomide; EHR: Electronic Health Records; JMDC: Japan Medical Center Data; THIN: The Health Improvement Network (UK); Optum: Optum de-identified Clinformatics Datamart (US); MDCR: ; IQVIA Disease Analyzer EMR (Germany); CCAE: IBM MarketScan Commercial Claims and Encounters (US); AMBEMR: IQVIA Ambulatory EMR (US)

Methods (II)

Propensity score (PS) stratification (quintiles) was undertaken and hazard ratios (HR) for MI and stroke were estimated for HCQ, SSZ and LEF compared to MTX in each dataset using Cox proportional-hazards models. Estimates were calibrated for residual confounding using negative control outcomes. Estimates were pooled where homogeneity across sources was adequate (I2<0.4). Intention to treat and an on treatment analysis were performed.

Results

165,782 RA patients initiating csDMARD therapy were included in the analyses (MTX: 73,996, HCL: 49,752, SSZ: 12,256, LEF: 9,244). The pooled incidence rate of MI and stroke for MTX was 7.64 and 10.26 per 1,000 person years. The pooled summary and source-specific estimated cHRs are shown in Figure 1 for the intention to treat analysis. Similar results were found for "on treatment" analyses.

Conclusions

Overall, all four csDMARDs had similar effects on MI risk. Interestingly, MTX was consistently associated with an increased risk of stroke compared to HCQ and SSZ. The observed risk differences among csDMARDs may be attributable to differential effect on the atherosclerotic process, differential overall disease control, or both.

This project has received funding from the Innovative Medicines Initiative 2 Joint Undertaking (JU) under grant agreement No 806968. The JU receives support from the European Union's Horizon 2020 research and innovation programme and EFPIA.